Economic Analysis of Short-Course Levofloxacin Versus Amoxicillin/Clavulanate in Treating Acute Bacterial Exacerbations of Chronic Bronchitis

Earnshaw SR,[1] Sikirica V,[2] Roskell N,[1] Shannon P,[1] Beard S,[1] Schein J,[2] Doshi D,[2] Janagap C[2]

[1]RTI Health Solutions, RTP, NC, USA

[2]Ortho-McNeil Janssen Scientific Affairs, LLC, Raritan, NJ, USA

BACKGROUND

- Chronic bronchitis has been estimated to occur in more than 12 million persons in the United States.[1]
- Costs of acute bacterial exacerbations of chronic bronchitis (ABECB) have been estimated at more than $1.2 billion for patients ≤65 years of age and >$419 million for patients over 65 years of age.[2]
- Failure to treat the infection with an antibiotic that can eradicate the pathogen in a timely manner can lead to increased likelihood of hospitalization and repeated courses of therapy.[3]
- The major goals of therapy for ABECB should be to provide rapid clinical resolution, eradicate the causative pathogen, and return respiratory function to pre-exacerbation baseline as quickly as possible.
- One standard treatment for treating adult outpatients with complicated ABECB is an administration of a 10-day course of amoxicillin/clavulanate.[4]
- Treatment with therapy that is just as effective and that has a similar impact on patient resource use and cost with shortened therapy duration can be advantageous.[5, 3]

OBJECTIVE AND PURPOSE

To examine the impact on costs and resource use of oral levofloxacin 750 mg (off-label) once-daily treatment for 5 days versus standard antibiotic treatment in adult outpatients with ABECB from a United States health care payer perspective.

CONTACT INFORMATION

Stephanie Earnshaw, PhD

RTI Health Solutions, RTI International
3040 Cornwallis Road, PO Box 12194
Research Triangle Park, NC 27709-2194

Phone: 919.485.2730
E-mail: searnshaw@rti.org

Presented at:
The Academy of Managed Care Pharmacy (AMCP)
19th Annual Meeting Showcase, San Diego, CA
April 11-14, 2007

METHODS

We conducted a post-hoc analysis of a randomized, multicenter, double-blind, parallel-group controlled trial (CAPSS-197) in adults with complicated ABECB (Figure 1).[5, 6]

- Complicated ABECB was defined as FEV1 <50% predicted, or FEV1 50% to 65% predicted and significant comorbidity or ≥4 exacerbations per year.
- Significant comorbidity was defined as diabetes mellitus, congestive heart failure, chronic renal or liver disease.
- Two different antibiotic regimens were compared and patients were followed up over a 45-day period:
 - Levofloxacin 750 mg orally once daily for 5 days, and
 - Amoxicillin/clavulanate 875/125 mg orally twice daily for 10 days.
- Safety and efficacy were evaluated in the intent-to-treat (ITT) and clinically evaluable populations.
- The ITT population was defined as all randomized patients. For ITT cost analyses, the population includes only subjects with health care utilization data.
- The clinically evaluable (CE) population was defined as all randomized subjects with the following characteristics[5]:
 - Had a confirmed diagnosis of ABECB;
 - Attended test-of-cure visit;
 - Took at least 80% and less than 120% of the protocol-specified study drug; and
 - Did not take any effective, concurrent, antibacterial agent from the day of enrollment in study until the post-therapy visit (i.e., during treatment), except when the subject was judged by the investigator to be a clinical failure.
- Cost analysis was performed in the ITT population and CE population.

Figure 1. Schematic Overview of the CAPSS-197 Trial

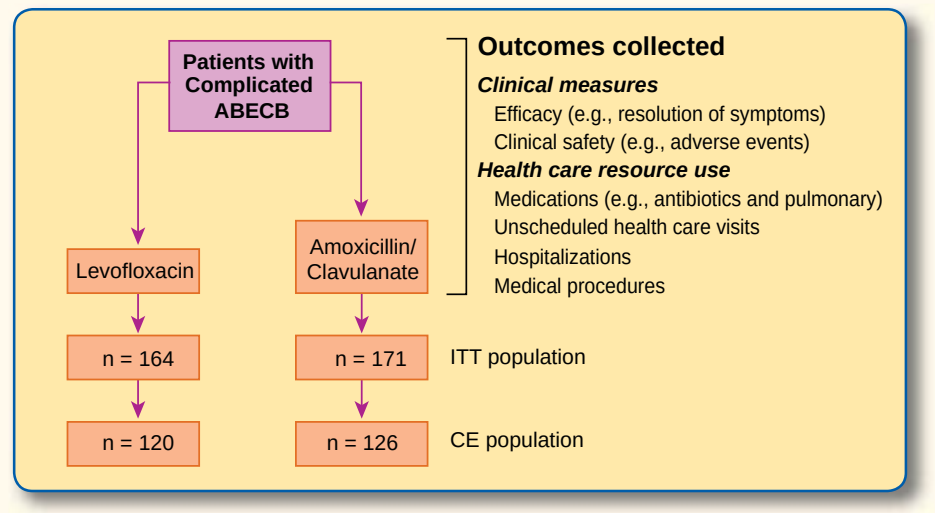
Patients with Complicated ABECB
Levofloxacin
Amoxicillin/ Clavulanate
n = 164
n = 171
ITT population
n = 120
n = 126
CE population
Outcomes collected
Clinical measures
Efficacy (e.g., resolution of symptoms)
Clinical safety (e.g., adverse events)
Health care resource use
Medications (e.g., antibiotics and pulmonary)
Unscheduled health care visits
Hospitalizations
Medical procedures

The following information was collected:

- Patient characteristics;
- Clinical success (cure or improvement) during on-therapy (Days 3-6), post-therapy, or test-of-cure (Days 17-26) and post-study visits (Days 40-45).
- Resource use was prospectively collected over a time period of up to 45 days:
 - Concurrent antibiotic medications;
 - Concomitant pulmonary medications (e.g., bronchodilators, oxygen, and cortisone);
 - Unscheduled health care visits, including outpatient medical visits and emergency room visits related to respiratory distress;
 - Days related to respiratory distress spent in a hospital, either in an intensive care unit or in a general medicine unit; and
 - Procedures used for diagnosis and treatment related to ABECB (e.g., bronchoscopy, chest x-ray, oxygen, laboratory tests, and other procedures).

Unit cost estimates were taken from the following sources:

- Drug costs: wholesale acquisition costs from Red Book[7];
- Outpatient visits and procedures: Medicare reimbursed costs from the Resource-Based Relative Value Scale[8]:
 - Current procedural terminology codes were assigned to each visit according to the reason for each visit and clinical opinion of severity when necessary.
- Hospitalization costs: Nationwide Inpatient Sample, Healthcare Cost, and Utilization Project database[9]:
 - Hospital charges were taken from a nationally representative sample of hospital discharges.
 - Charges are converted to costs using published cost-to-charge ratios.[10]
 - Costs were adjusted to 2005 United States dollars using the medical component of the consumer price index.[11]

Statistical analysis consisted of the following:

- Descriptive statistics of resource use and costs:
 - Total number and percentage of patients experiencing the event;
 - Mean number of resources used per patient;
 - Number of concurrent antibiotic and pulmonary medications prescribed; and
 - Mean cost per patient.
- Bootstrap analysis:
 - Cost analyses were conducted with branded costs of medications, and generic costs were reviewed.
 - Uncertainty in the estimate of the difference in the mean cost between treatment groups was assessed by constructing a bootstrap (95% CI).
 - Iterative unrestricted random samplings (with replacement) from study population (10,000 iterations) were performed.
 - Bootstrap mean difference in total cost is the mean of these 10,000 estimates. A 95% confidence interval was derived using 2.5% and 97.5% percentiles.

RESULTS

Table 1. Patient Characteristics of the Intent-to-Treat Population

Characteristic	Levofloxacin (n = 164) n (%)	Amoxicillin/ Clavulanate (n = 171) n (%)	p-value
Demographic			
Female	75 (45.7)	89 (52.0)	0.25 (χ^2)
Caucasian	127 (77.4)	131 (76.6)	0.86 (χ^2)
Age (Mean)	59.3±14.2	60.2±12.6	0.54 (t-test)
Age range (Years)	18-91	20-85	
Current smokers	76 (46.3)	85 (49.7)	0.74 (χ^2)
Ex-smoker	74 (45.1)	70 (40.9)	
Nonsmoker	14 (8.5)	16 (9.4)	
Exacerbation Frequency in Past 12 Months			
0	7 (4.3)	5 (2.9)	0.65 (χ^2)
1–3	63 (38.4	55 (32.2)	
4–6	84 (51.2)	101 (59.1)	
7–9	5 (3.0)	6 (3.5)	
10+	5 (3.0)	4 (2.3)	
Exhibits Significant Comorbidity			
	74 (45.1)	78 (45.6)	0.93 (χ^2)

No statistically significant differences occurred in the clinically evaluable population patient characteristics.

Table 2. Clinical Efficacy Between Treatment Groups

Clinical Success	Levofloxacin n (%)	Amoxicillin/Clavulanate n (%)	95% CI
Intent-to-Treat Population	n = 164	n = 171	
Clinical Success, Test-of-Cure Visit	121 (73.78%)	137 (80.12%)	-2.67, 15.34
Clinical Success, Post-Study Visit	89 (54.27%)	96 (56.14%)	-8.78, 12.52
Clinically Evaluable Population	n = 120	n = 126	
Clinical Success, Test-of-Cure Visit	95 (79.17%)	103 (81.75%)	-7.34, 12.49
Clinical Success, Post-Study Visit	74 (61.67%)	81 (64.29%)	-9.45, 14.69

Analysis results remain robust with varying hospitalization, branded, or generic costs of medications.

Table 3. Bootstrapped Confidence Intervals on Total Cost Difference

Population	Mean Difference in Total Cost*	Bootstrapped Mean Difference in Total Cost	Bootstrapped 95% CI
Intent-to-Treat	$-173.11	$-172.26	$-524.20, $156.16
Clinically Evaluable	$-225.54	$-227.74	$-532.93, $67.36

Proportion of patients with zero additional costs in the ITT (23.8% and 19.9%) and CE population (25.0% and 19.8%) were similar between levofloxacin and amoxicillin/clavulanate-treated patients.

*Mean difference is levofloxacin minus amoxicillin/clavulanate.

Figure 2. Comparison of Average Costs Over 45 Days by Cost Category in the Intent-to-Treat Population

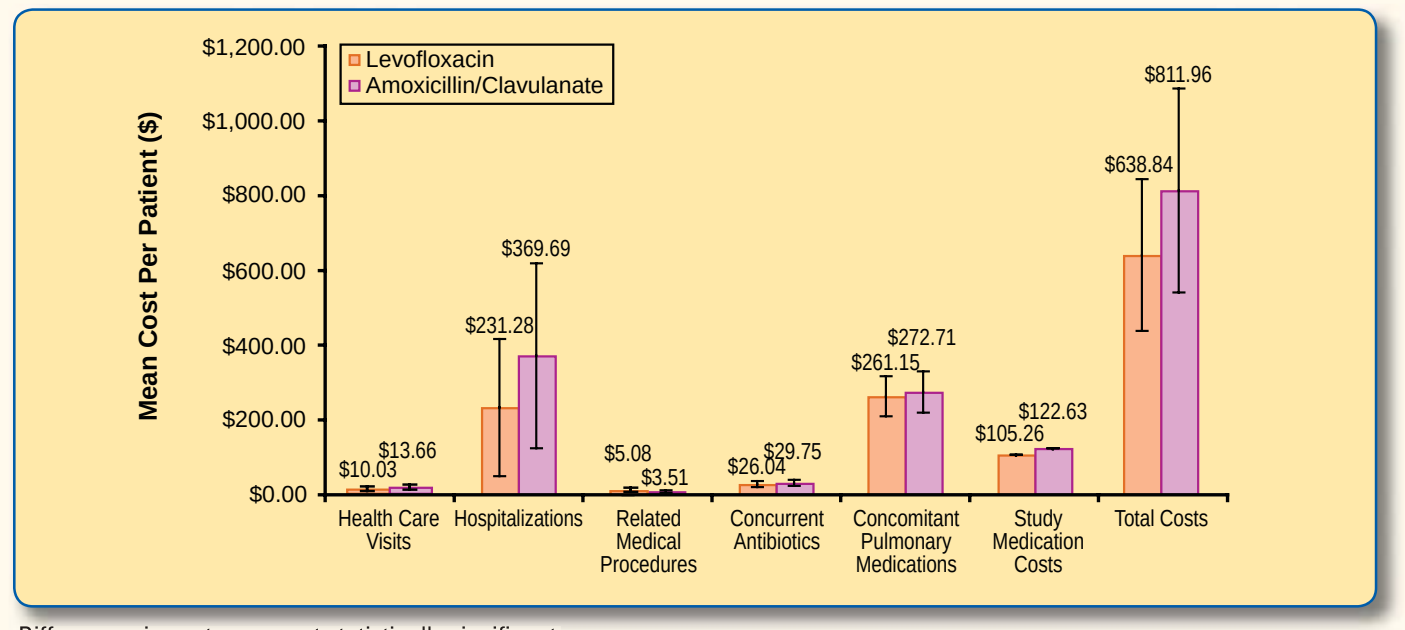
Mean Cost Per Patient ($)
$1,200.00
$1,000.00
$800.00
$600.00
$400.00
$200.00
$0.00
Levofloxacin
Amoxicillin/Clavulanate
$10.03
$13.66
$231.28
$369.69
$5.08
$3.51
$26.04
$29.75
$261.15
$272.71
$105.26
$122.63
$638.84
$811.96
Health Care Visits
Hospitalizations
Related Medical Procedures
Concurrent Antibiotics
Concomitant Pulmonary Medications
Study Medication Costs
Total Costs

Differences in costs were not statistically significant.

Figure 3. Differences in Costs Over 45 Days by Cost Category in the Intent-to-Treat Population

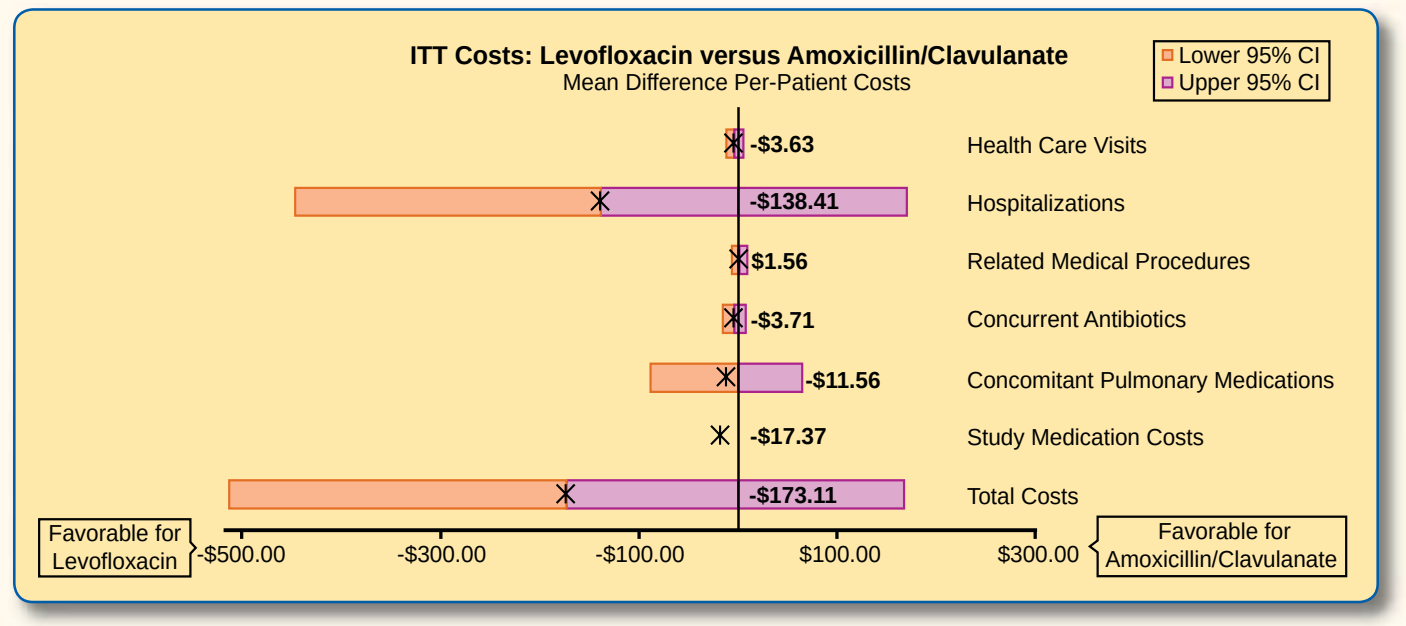
ITT Costs: Levofloxacin versus Amoxicillin/Clavulanate
Mean Difference Per-Patient Costs
Lower 95% CI
Upper 95% CI
-$3.63
Health Care Visits
-$138.41
Hospitalizations
$1.56
Related Medical Procedures
-$3.71
Concurrent Antibiotics
-$11.56
Concomitant Pulmonary Medications
-$17.37
Study Medication Costs
-$173.11
Total Costs
Favorable for Levofloxacin
-$500.00
-$300.00
-$100.00
$100.00
$300.00
Favorable for Amoxicillin/Clavulanate

Figure 4. Comparison of Average Utilization Over 45 Days by Category in the Intent-to-Treat Population

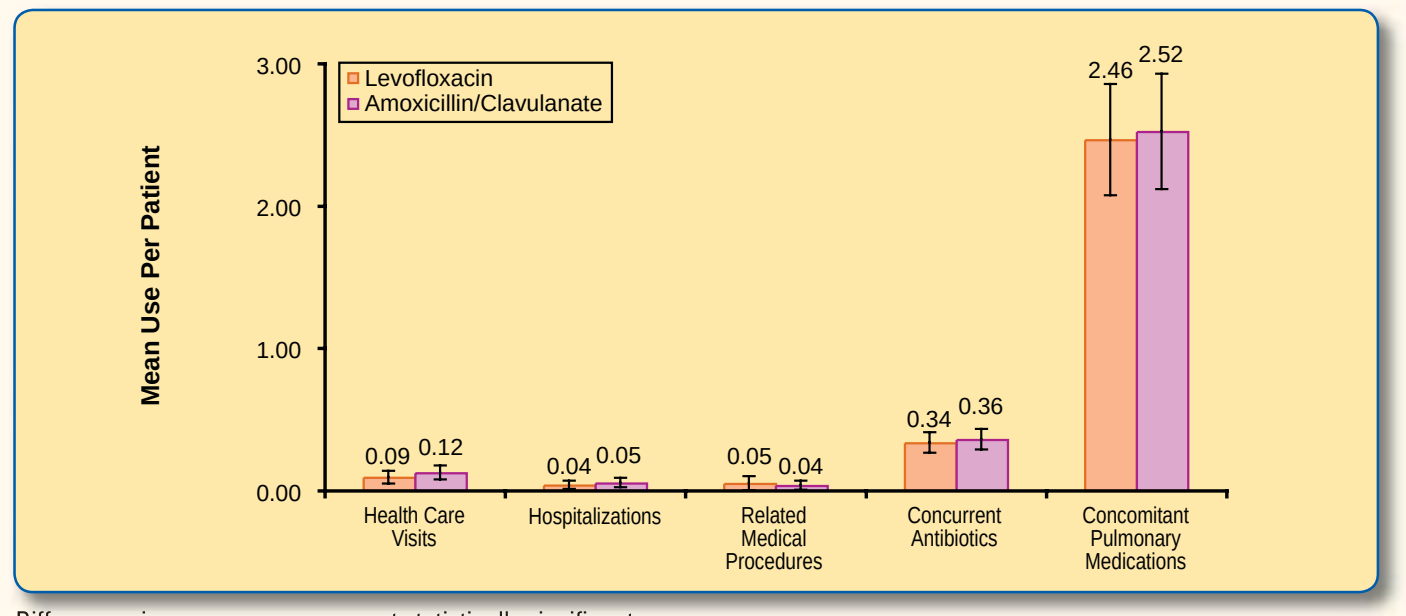
Mean Use Per Patient
3.00
2.00
1.00
0.00
Levofloxacin
Amoxicillin/Clavulanate
0.09
0.12
0.04
0.05
0.05
0.04
0.34
0.36
2.46
2.52
Health Care Visits
Hospitalizations
Related Medical Procedures
Concurrent Antibiotics
Concomitant Pulmonary Medications

Differences in resource use were not statistically significant.

Figure 5. Differences in Utilization Over 45 Days by Category in the Intent-to-Treat Population

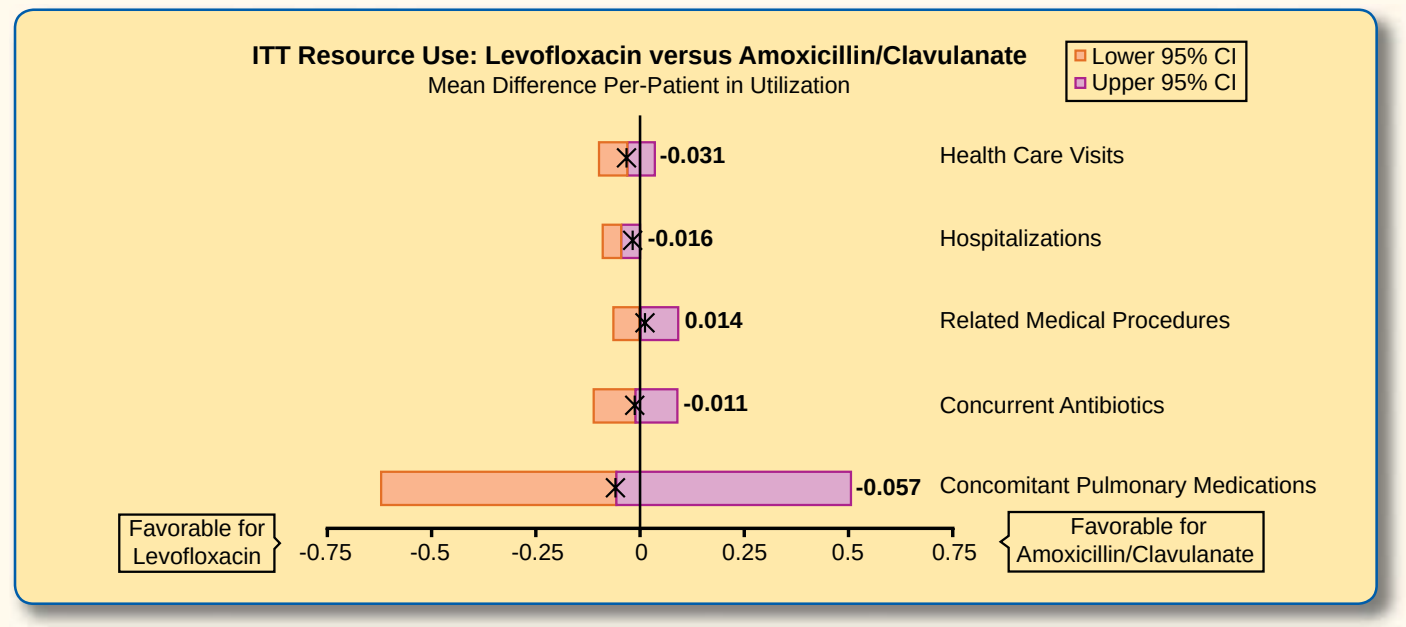
ITT Resource Use: Levofloxacin versus Amoxicillin/Clavulanate
Mean Difference Per-Patient in Utilization
Lower 95% CI
Upper 95% CI
-0.031
Health Care Visits
-0.016
Hospitalizations
0.014
Related Medical Procedures
-0.011
Concurrent Antibiotics
-0.057
Concomitant Pulmonary Medications
Favorable for Levofloxacin
-0.75
-0.5
-0.25
0
0.25
0.5
0.75
Favorable for Amoxicillin/Clavulanate

LIMITATIONS

- The cost application study was conducted using data from a clinical trial. Therefore, results may not be generalizable to a real-world setting.
- Since there is limited information in the public domain regarding the use and cost of levofloxacin 750 mg for ABECB, we chose the most appropriate population from the study (ITT) to represent real-world use.

CONCLUSIONS

- Levofloxacin and amoxicillin/ clavulanate showed similar efficacy and safety.
- We found no significant differences in costs when comparing a 5-day course of levofloxacin and a 10-day course of amoxicillin/clavulanate.
- This study was adequately powered to reach meaningful conclusions for clinical outcomes. It did not have sufficient power to assess economic outcomes. Adequately sized studies will need to be done.

REFERENCES

1. American Thoracic Society [ATS]. Am J Respir Crit Care Med. 1995 Nov;152(5 Pt 2):S77-121.
2. Niederman MS, McCombs JS, Unger AN, Kumar A, Popovian R. Clin Ther 1999;21(3):576-91.
3. Grossman RF. Chest 1997;112:310S-3S.
4. Asia Pacific COPD Roundtable Group. Respirology. 2005 Jan;10(1):9-17. Review.
5. Martinez FJ, Grossman RF, Zadeikis N, Fisher AC, Walker K, Ambruzs ME, et al. Eur Respir J 2005;25:1001-10.
6. CAPSS-197 Report. 08 December 2003. Data on file.
7. Red Book. Montvale: Thomas PDR; 2005.
8. Medicare RBRVS. Chicago: American Medical Association; 2005.
9. Agency for Healthcare Research and Quality. Rockville, MD; 2002.
10. Centers for Medicare and Medicaid Services [CMS]. Available at: http://www.cms.hhs.gov/MedicareFeeforSvcPartsAB/03_MEDPAR.asp. Accessed Mar 8, 2002.
11. Medical Consumer Price Index. 2005. Available at: http://www.grsnet.com/grs_library/index.html. Accessed December 22, 2006.