Utility of Multiple Databases in a Surveillance Program for Prescription Narcotic Analgesics

Kirk Midkiff, MPH[1]; Catherine Johannes, PhD[1]; Aparna Mohan, MD, PhD[2]

[1]RTI Health Solutions, Research Triangle Park, NC, United States

[2]Johnson & Johnson Pharmaceutical Research & Development, Titusville, NJ, United States

ABSTRACT

Background: The surveillance of possible abuse and misuse of products is important in pharmacovigilance programs for prescription narcotic analgesics, but information needed may not be readily available from a single data source.

Objectives: To describe how data from two different sources were used to monitor overmedication and nonmedical use of 1) fentanyl-containing products as a group and 2) all prescription narcotics in an ongoing company-sponsored safety surveillance program.

Methods: We obtained emergency department (ED) reports of overmedication and nonmedical use of the drug groups of interest from the Drug Abuse Warning Network (DAWN). Denominators were estimated by obtaining population exposure data from a prescription retail pharmacy database from IMS Health. We estimated event rates among people dispensed a narcotic using the number of DAWN ED visits for overmedication and nonmedical use during 2004-2005 as the numerator and the projected number of patients dispensed a narcotic of interest as the denominator. We used the delta method to obtain variance estimates for calculation of 95% confidence intervals (CI).

Results: About 70 million people had exposure to any prescription narcotic analgesic in each year from 2004-2006; of these, about 1.3 million people each year had a fentanyl exposure. For all prescription narcotic analgesics the crude ED visit rates in 2004 and 2005 of overmedication were 70 (95% CI: 54-90) and 89 (95% CI: 69-114), and of nonmedical use were 116 (95%CI: 95-141) and 143 (95% CI: 113-182). The corresponding rates for all fentanyl products were 205 (95% CI: 136-309) and 368 (95% CI: 261-519) for overmedication and 420 (95% CI: 303-582) and 345 (95% CI: 244-487) for nonmedical use.

Conclusions: These visit rates have limited usefulness as stand-alone point estimates owing to the small number of cases and large variances. These rates can, however, provide information on time trends in the possible misuse and abuse of prescription narcotic analgesics, aiding in signal detection.

BACKGROUND

- Surveillance of possible abuse and misuse of a product is an important component of pharmacovigilance programs for prescription narcotic analgesics.
- It is difficult to obtain prescribing data and data on narcotic misuse and abuse from a single data source.
- Specialized data sources, such as DAWN, can provide data on misuse and abuse; other data sources, such as prescription claims databases, must be used to obtain denominator information on drug utilization.
- DAWN captures data from medical records of drug- and alcohol-related ED visits and provides data weighted to the United States (US) population (by age and gender) for different types of visits.
- The Longitudinal Prescription Database (LRx), operated by IMS Health, Inc., captures data on individual patients from retail pharmacy transactions in the US and can be used to provide data on the total number of patients exposed to various products and product groupings.

OBJECTIVE

This study was designed to describe how data from two different sources were used to monitor overmedication and nonmedical use of (1) fentanyl-containing products as a group; and (2) all prescription narcotics in an ongoing Company-sponsored safety surveillance program.

METHODS

Data Sources

DAWN

DAWN is an active public health surveillance system administered by the Substance Abuse and Mental Health Services Administration (SAMHSA). DAWN collects information on drug-related ED visits and drug-related deaths from a national sample of approximately 500 hospital EDs.[1] SAMHSA publishes weighted summary-level estimates annually. Weighted estimates are representative of the entire US. Manufacturers may obtain weighted estimates on specific narcotic agents upon special request to SAMHSA.

Case Definition and Ascertainment

- Cases are identified through retrospective review of all medical records in each hospital in the DAWN system. An ED visit becomes a DAWN case if, based on clinician documentation, the visit is judged to be for a condition induced by or related to recent drug use.
- Cases are classified into one of eight case types (i.e., suicide attempt, seeking detoxification, alcohol only in patients under age 21, adverse reaction, overmedication, malicious poisoning, accidental ingestion, and other).

DAWN Data

- DAWN data, weighted to the US population to provide national estimates of ED visits, are provided to the requestor by SAMHSA in the form of populated data tables.

LRx

LRx is a longitudinal de-identified, record-linked prescription database containing information on patient demographics, retail prescription activity, and prescribing detail. LRx captures approximately 50% of all retail transactions in the US, representing data assembled from chain and independent retail pharmacies, mass merchants, grocers, and system vendors. Data captured are from more than 150 million unique patients and 1 million prescribers. Projection factors, based on product and region, are developed from a separate database containing data for total prescription volume (Xponent) and applied to a patient sample in the LRx database to estimate the total number of patients exposed to a product or product grouping in the retail chain.

LRx Data

- LRx data, projected to the US population to provide national estimates of patients exposed to certain products or product groupings, are provided to the requestor by IMS Health in the form of populated data tables.

Reporting Sources

SAMHSA's Office of Applied Studies (OAS) provided national estimates from DAWN of the number of ED visits following a special request by the manufacturer.

IMS Health provided national estimates of the number of patients exposed to the drug groupings of interest.

Definition of Variables

- Two ED visit types were included in this analysis:
 - Overmedication: An ED visit where nonmedical use, overuse, or misuse of prescription narcotic analgesics is indicated in the medical record, but it is not documented as drug abuse.
 - Nonmedical use: A DAWN visit that has been classified as "other" case type. "Other" case type is an ED visit related to a prescription narcotic analgesic that could not be assigned to any of the other seven possible case types in DAWN.
- Exposure variables: All narcotic analgesics and all fentanyl products, including transdermal and nontransdermal systems.
- ED event rate: Calculated as (Number of Estimated DAWN ED Visits Involving Drug Group of Interest During Time T) / (Number of Patients Dispensed Drug Group of Interest During Time T).

Study Design and Analysis

- We obtained tabulations of DAWN ED visit estimates for overmedication and nonmedical use reported for fentanyl-containing products as a group, and separately for all prescription narcotic analgesics in the US, for 2004 and 2005 (data for 2006 were not available at the time of analysis).
- We obtained population exposure data for fentanyl-containing products as a group, and separately for all patients dispensed a prescription narcotic analgesic in the US, for 2004, 2005, and 2006.
- We calculated crude incidence event rates by year for the DAWN events of overmedication and nonmedical use per 100,000 person-years for those dispensed a drug grouping of interest.
- We used the delta method[2] to obtain variance estimates for calculating the 95% CIs for the event rates using the following formula and assumptions:
 - The delta method was used to prepare the variance estimator for the ratio of two random variables x and y (Equation 1). The numerator and denominator are expressed as random variables x and y, both having expected values expressed as E(x) and E(y). The derived equation and underlying assumptions are expressed as follows:
- Assumptions
 - The distribution of r will be asymmetrical because it is a ratio measure; therefore, logarithmic transformation of r (Equation 2) was used.
 - Equation 3 is an approximation of the variance, which assumes the variance is known for x and y, expressed as Var(x) and Var(y), respectively; random variables x and y are independent.
 - Because the numerator and denominator are estimates, as well as the standard error associated with each, Equation 4 must be used to estimate the variance for the ratio of the logarithm of the two random variables. Once the estimated variance is calculated, Equation 5 is used to calculate the upper and lower bounds of the 95% CI.

Equation 1 $$r = \frac{x}{y}$$

Equation 2 $$\ln[r] = \ln\left[\frac{x}{y}\right]$$

Equation 3 $$Var(\ln[r]) = \left(\frac{Var(x)}{\mu_x^2} + \frac{Var(y)}{\mu_y^2}\right)$$

Equation 4 $$\hat{Var}(\ln[r]) = \left(\frac{\hat{Var}(x)}{\mu_x^2} + \frac{\hat{Var}(y)}{\mu_y^2}\right)$$

For calculations 1, 2, 3, and 4:

$$r = \ln\left[\frac{\hat{\mu}_x}{\hat{\mu}_x}\right]$$

Equation 5 $$CI(95\%) = \exp\left[\ln[\hat{r}] \pm 1.96\sqrt{\hat{Var}(\ln[r])}\right]$$

RESULTS

Table 1. National Estimates of Number of People Exposed, by Product Grouping and Period of Interest[a]

Period	Opioid Product Category[b]	
	All Fentanyl Products	All Narcotic Analgesics
2004	1,280,393	71,239,975
2005	1,284,166	68,892,410
2006	1,326,995	70,309,917

[a] Includes all patients using the products mentioned in the table during the reporting period (e.g., new, add-ons, switches, continuation). The table is indexed at the patient level longitudinally. A patient is counted on the drug group of interest only once during a given period regardless of length on therapy during the period or the number of prescriptions written during the period. Patients whose therapy spans 2 periods are counted as exposed in both periods.

[b] All pharmaceutical opioids, which excludes illicit opioids (e.g., heroin).

- Approximately 70 million people had exposure to any prescription narcotic analgesic in each year from 2004 to 2006 (Table 1); of these, approximately 1.3 million people each year had a fentanyl exposure.

Emergency Department Visit Rates

- ED visit rates for overmedication from all narcotic analgesics increased from 70 visits (in 2004) to 89 visits (in 2005) per 100,000 person-years (Figure 1).
- ED visit rates for nonmedical use from all narcotic analgesics increased from 116 visits (in 2004) to 143 visits (in 2005) per 100,000 person-years (Figure 1).
- ED visit rates for overmedication and nonmedical use related to all fentanyl products moved in opposite directions from 2004 to 2005, with overmedication moving up and nonmedical use moving down year to year (Figure 1); 2006 event data were not available to evaluate additional evidence on time trends.
- Confidence intervals (CIs) for all calculated rates were wide (Figure 1).

Figure 1. DAWN Emergency Department Visit Rates[a] and 95% CI[b] by Event Type, Product,[c] and Year

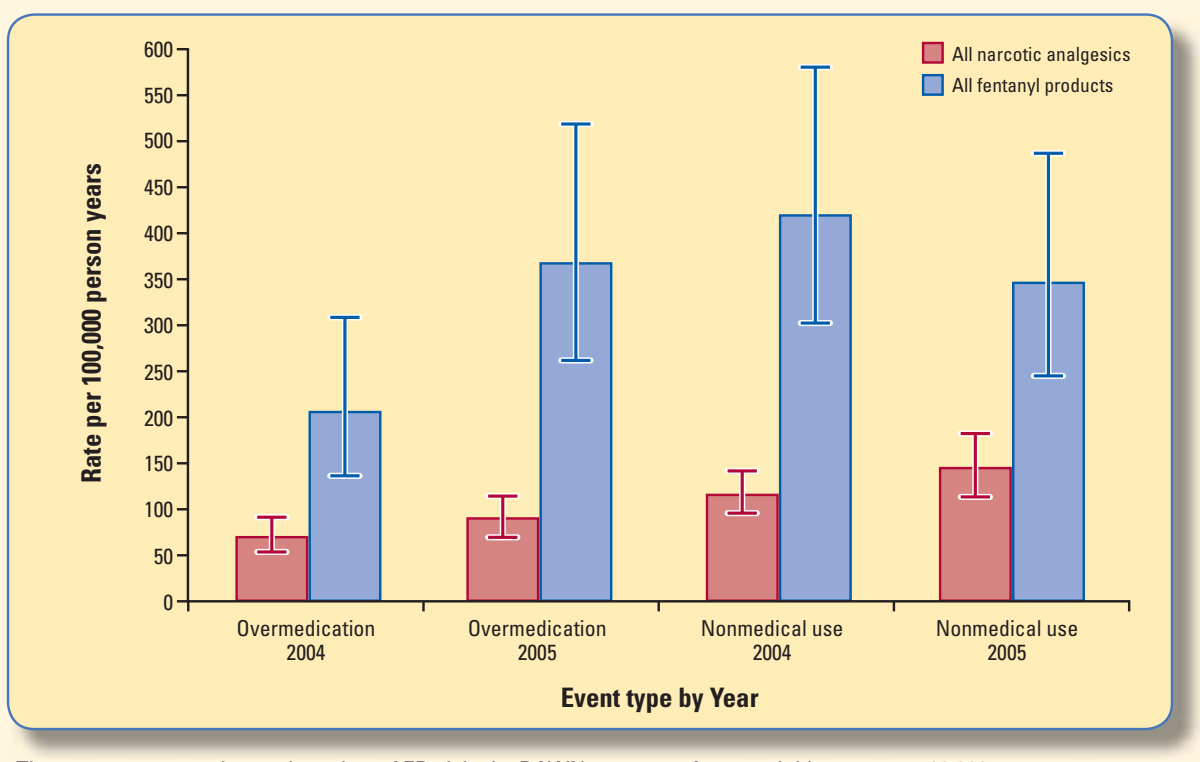
All narcotic analgesics
All fentanyl products
Rate per 100,000 person years
600
550
500
450
400
350
300
250
200
150
100
50
0
Overmedication 2004
Overmedication 2005
Nonmedical use 2004
Nonmedical use 2005
Event type by Year

[a] The rates represent the total number of ED visits by DAWN case type for an opioid group per 100,000 persons-years exposed. The rates assume exposure for the entire year for each person.

[b] CI is represented by a vertical bracket.

[c] All pharmaceutical opioids, which excludes illicit opioids (e.g., heroin).

CONCLUSIONS

- When data are collected from two independent sources, a method, such as the delta method, is needed to calculate a combined variance for preparing CIs; however, the numerator and the denominator must be independent random variables.
- The ED visit rates calculated using the two different sources (DAWN data and national prescribing data) have limited usefulness as stand-alone point estimates but can provide information on possible time trends in narcotic misuse as more years of data become available.
- Such surveillance activities can aid in signal detection efforts for marketed narcotic products.

REFERENCES

1. Substance Abuse and Mental Health Services Administration, Office of Applied Studies [SAMHSA]. Drug Abuse Warning Network, 2004. National estimates of drug related emergency department visits. DAWN Series D-28. Publication (SMA) 06-4143. Rockville, MD; 2006.
2. Bishop YM, Fienberg SE, Holland PW. Discrete multivariate analysis. Cambridge, MA: Massachusetts Institute of Technology Press; 1975.

CONFLICT OF INTEREST:

KM and CJ are employees of RTI Health Solutions (an independent nonprofit research organization that does work for government agencies and pharmaceutical companies), and AM is an employee of Johnson & Johnson Pharmaceutical Research & Development, LLC.

CONTACT INFORMATION

Kirk Midkiff, MPH

RTI Health Solutions, RTI International
200 Park Offices Drive
Research Triangle Park, NC 27709

Phone: +1.919.541.6638
Fax: +1.919.541.7222

E-mail: kmidkiff@rti.org

Presented at: 24th International Conference on Pharmacoepidemiology and Therapeutic Risk Management

August 17-20, 2008,

Copenhagen, Denmark