PHP97

PRO label claims: an analysis based on a review of PROs among NMEs and BLAs 2006–2010

Mordin M[1], Clark M[1], DeMuro C[2], Evans E[2], Copley-Merriman K[1], Fehnel SE[2], Gnanasakthy A[3]

[1]RTI Health Solutions, Ann Arbor, MI, USA; [2]RTI Health Solutions, Research Triangle Park, NC, USA; [3]Novartis Pharmaceuticals Corporation, East Hanover, NJ, USA

ABSTRACT

Background/Objective: Wilke and colleagues (2004) previously conducted a review of effectiveness endpoints reported in the labels of new drug products approved in the United States (US) between 1997-2002 to determine the extent and type of PRO endpoints utilized. They reported that 30% of product labels reviewed included PROs. Our study aimed to build upon this work by describing the current state of PRO label claims granted for new molecular entities (NMEs) and biologic license applications (BLAs) following release of the draft and final FDA PRO Guidance documents (i.e., since February 2006).

Methods: Using the FDA Drug Approval Reports webpage, all FDA approved NMEs and BLAs between January 2006 and December 2010 were identified. Generic products with tentative approvals granted in this period were excluded. For all identified drug products, medical review sections from publicly available drug approval packages (DAP) were reviewed to identify PRO endpoint status. Product labels (indication, clinical trials sections) were reviewed to determine the number and type of PRO claims.

Results: Of the 116 NMEs/BLAs identified, 28 (24.1%) were granted PRO claims. The majority (n=24) were for signs and symptoms. Nine of the signs and symptom claims were pain-related. Of the 28 products with PRO claims, a PRO was a primary endpoint for 20 (71%). All 20 of these primary endpoints were symptom-related and the majority (12 of 20) were collected via diary.

Conclusions: PRO claims continue to be approved by FDA, with 24% of NMEs and BLAs granted PRO claims. Successful PRO label claims over the past five years have been largely in support of treatment benefit for symptoms specified as primary endpoints. The proportion of NMEs with PRO label claims during the post-guidance period (24.1%) was lower than that of the pre-guidance period (30%).

BACKGROUND

- Content of package inserts (PIs) from the United States (US) Food and Drug Administration (FDA) is vital to the commercial success of a medicinal product
- PRO use is particularly common from products developed to treat chronic, disabling conditions where the intention is not necessarily to cure but to ameliorate symptoms, facilitate functioning, or improve quality of life
- The FDA's release of the Draft (2006) and Final (2009) Guidance for Industry Patient Reported Outcomes: Use in Medical Product Development to Support Labeling Claims[1,2] (PRO Guidance) were landmark events
- In 2004, Willke and colleagues conducted a review of effectiveness endpoints reported in the labels of new drug products approved in the United States from 1997 through 2002 to determine the extent and type of patient-reported outcome (PRO) endpoints used[3]
- They reported that 30% of product labels reviewed included PROs[3]

OBJECTIVES

- Our study aimed to describe the current state of PRO label claims granted for new molecular entities (NMEs) and biologic license applications (BLAs) following release of the Draft and Final FDA PRO Guidance documents

METHODS

- The FDA Drug Approval Reports Webpage was used to determine the number of products approved in the US from January 2006 through December 2010. Original New Drug Approvals (NDAs) and BLAs by month were selected. The reports include specification of Center for Drug Evaluation Research (CDER) NDA chemical classification. Our review included products classified by CDER as NMEs or BLAs
- Drug approval packages (DAP) and approved product labels were reviewed for each product. As available, information was retrieved from the medical review, summary review, cross-discipline team leader review, and other review sections from the DAP, as well as the Indication and Clinical Studies section of the approved product label. As available, the following information was collected for each US drug product identified:
 - Brand name
 - Generic name
 - Date of approval
 - Applicant
 - Label indication
 - PRO claim language
 - PRO instruments named in label
 - Type of PRO claim (yes/no)
 - Signs and symptoms
 - Functioning
 - HRQOL
 - Patient global rating
 - Other
 - Reviewing division
 - Medical review available (yes/no)
 - Indication in DAP of Study Endpoints and Label Development (SEALD) review (yes/no)
 - PRO measures mentioned in the label and DAP, and endpoint status (primary, secondary, tertiary/exploratory)
 - PRO results reported as statistically significant (yes/no)

Statistical analysis consisted of frequencies and cross-tabulations of measured characteristics. Calculations were performed using Microsoft Excel 2007.

RESULTS

- 116 NMEs/BLAs were identified
- 28 products (24.1%) were granted 38 PRO label claims
- The majority (n=24) were for signs and symptoms (Table 1)
 - Nine of the signs and symptom claims were pain-related

Table 1 Types of Claims Granted

Type of Claim	Products with PRO Claim (N=28)	
	N	%
Signs & Symptoms	24	85.70%
Functioning	7	25.00%
HRQOL	2	7.10%
Patient Global Rating	3	10.70%
Other	2	7.10%

- Of the 28 products with PRO claims, a PRO was a primary endpoint for 20 (71%) (Table 2)

Table 2 PRO Primary Endpoint and Signs and Symptoms Claims

	PRO Primary Endpoint		TOTAL # Products
	Yes	No	
Signs & Symptom Claim: Yes	20	4	24
Signs & Symptom Claim: No	0	4	4
Total	20	8	28

- All 20 of these primary endpoints were symptom related and the majority (12 of 20) were collected via diary. Table 3 lists the PRO measures for these products
- Table 4 presents the PRO measures used for the 8 products that received PRO claims but where PRO was not a primary endpoint
- Among the 28 products with PRO claims in the label, the FDA Divisions of Neurology Products (n=7; 25.0%) and Anesthesia, Analgesia, and Rheumatology Products (n=6; 21.4%) granted the most PRO label claims (Table 5). See related poster PHP96 on reasons for rejection of PRO claims.

Table 3 Products with PRO Label Claims and PRO was a Primary Endpoint (n=20)

Product	PRO Measure Included
AZILECT	Three registration trials (TEMPO, PRESTO, LARGO) TEMPO: Unified Parkinson's Disease Rating Scale (UPDRS) PRESTO/LARGO: Total daily "off" time recorded in patient diaries
OMNARIS	Reflective Total Nasal Symptom Score (rTNSS)
ARCALYST	Daily Health Assessment Form with a 0–10 scale for each symptom (assessing signs and symptoms of the disease)
CIMZIA	Crohn's Disease Activity Index (CDAI) (clinician-administered PRO assessing disease symptoms)
TOVIAZ	Urge urinary incontinence episodes per 24 hours and number of micturitions (frequency) per 24 hours
RAPAFLO	International Prostate Symptom Score (IPSS) which evaluates irritative (frequency, urgency, and nocturia), and obstructive (hesitancy, incomplete emptying, intermittency, and weak stream) symptoms
VIMPAT	Subject diary recording of seizure frequency
BANZEL	Seizure severity from the Parent/Guardian Global Evaluation of the patient's condition (proxy PRO)
NUCYNTA	Pain via an 11-point rating scale ranging from 0 to 10
SAVELLA	100mm PI-VAS, Patient Global Impression of Change SF-36 Physical Component Score (PCS)
DYSPORT	Cervical dystonia indication: TWSTRS Glabellar Lines indication: Patient Global Assessment of Change in Glabellar Line Severity
SIMPONI	RA and PsA Indications: Health Assessment Questionnaire Disability Index (HAQ-DI) Patient's assessment of pain (0-10) AnkSpon Indication: Total back pain (0-10), Inflammation (mean of two patient-reported stiffness self-assessments in the BASFI)
CETIRIZINE-ALLERGY	Symptoms of runny nose, itchy, watery eyes, sneezing; itchy nose
CETIRIZINE-HIVES	Itching
SABRIL	CPS seizure frequency
BEPREVE	Ocular itching at 3, 5, and 7 minutes post CAC and investigator-evaluated conjunctival redness at 7, 15, and 20 minutes post CAC. Itching and redness scales were based on a 5-unit (9 steps) grading scale with half unit (one step) increments allowed
KALBITOR	Mean Symptom Complex (MSCS) and Treatment Outcome Score (TOS)
ACTERMA	ACR Response (Pain VAS, Patient Global Assessment, HAQ-DI)
XEOMIN	TWSTRS
LASTACAFT	Ocular itching evaluated by the subject at 3, 5, and 7 minutes post-challenge, measured on a 0-4 unit scale

We note several hybrid measures that combined both clinician-reported outcomes (ClinROs) and PROs into a single measurement tool. While these hybrid measures are not solely patient-reported, they contain PROs that are critical to assessing efficacy in the given indications.

Table 4 Products with PRO label claims and PRO was not a Primary Endpoint (n=8)

Product	PRO Measures Included
VYVANASE	Conners' Parent Rating Scale
CHANTIX	Brief Questionnaire of Smoking Urges Minnesota Nicotine Withdrawal scale Smoking Effects Inventory
SOLIRIS	FACIT-Fatigue EORTC QLQ-C30
DUREZOL	Visual Analog Scale (VAS) - eye pain/discomfort VAS - photophobia
AMPYRA	12-item Multiple Sclerosis Walking Scale (MSWS-12) (patient self-assessment of ambulatory disability)
LETAIRIS	SF-36 Health Survey
ASCLERA	Patient satisfaction using a verbal rating scale
EGRIFTA	Distress associated with belly appearance

Table 5 Reviewing Division Statistics

Reviewing Division	Number of products approved	Number of Products with PRO Claim	Type of Claim Granted					
			Signs & Symptoms	Functioning	HRQOL	Patient Global Rating	Other	Total
Anesthesia, Analgesia and Rheumatology Products	10	6	6	3	0	2	0	11
Antimocrobial Products	1	1	1	0	0	0	0	1
Anti-infective and Ophthalmology Products	8	2	2	0	0	0	0	2
Antiviral Products	8	0	0	0	0	0	0	0
Biologic Oncology Products	2	0	0	0	0	0	0	0
Cardiovascular and Renal Products	10	2	0	0	1	0	1	2
Dermatology and Dental Products	3	0	0	0	0	0	0	0
Drug Oncology Products	16	0	0	0	0	0	0	0
Gastroenterology Products	8	1	1	0	0	0	0	1
Medical Imaging and Hematology Products	9	1	1	0	1	0	0	2
Metabolism and Endocrinology Products	7	1	0	0	0	0	1	1
Neurology Products	11	7	6	4	0	1	0	11
Nonprescription Clinical Evaluation	3	2	2	0	0	0	0	2
Psychiatry Products	7	1	1	0	0	0	0	1
Pulmonary and Allergy Products	3	2	2	0	0	0	0	2
Reproductive and Urologic Products	5	2	2	0	0	0	0	2
Special Pathogen and Transplant Products	5	0	0	0	0	0	0	0
Total	**116**	**28**	**24**	**7**	**2**	**3**	**2**	**38**

CONCLUSIONS

- PRO claims continue to be approved by FDA, with 24% of NMEs and BLAs granted PRO claims
- Successful PRO label claims over the past five years have been largely in support of treatment benefit for symptoms specified as primary endpoints
- The proportion of NMEs with PRO label claims during the post-guidance period (24.1%) was slightly lower than that of the pre-guidance period (30%)

LIMITATIONS

- This review was limited to products with NDA Chemical Type classified as NME or BLA. Products undergoing FDA review as a new formulation, new combination or new indication were not reviewed. The reviews for these classifications may result in a different proportion of products granted PRO label claims
- This review is limited to the information that is publicly available on the FDA website; undocumented informal consultations or conversations between FDA Reviewing Divisions and sponsors cannot be ruled out
- The date of the final drug product approval may reflect months or years of regulatory interaction

REFERENCES

1. US Department of Health and Human Services. Draft Guidance for industry. Patient-reported outcome measures: use in medical product development to support labeling claims. February 2006.
2. US Department of Health and Human Services. Guidance for industry. Patient-reported outcome measures: use in medical product development to support labeling claims. December 2009. Available at: http://www.fda.gov/downloads/Drugs/GuidanceComplianceRegulatoryInformation/Guidances/UCM193282.pdf. Accessed January 14, 2011.
3. Willke RJ, Burke LB, Erickson P. Measuring treatment impact: a review of patient-reported outcomes and other efficacy endpoints in approved product labels. Control Clin Trials. 2004 Dec;25(6):535–52.

Poster presented at ISPOR 16th Annual International Meeting, May 21–25, 2011, Hilton Baltimore Baltimore, MD, USA