Inpatient Costs and Outcomes Associated With Traumatic Injury in the United States by Injury Severity and Trauma Center Designation

Candrilli SD,[1] Davis KL,[1] Mitra D,[1] Tortella BJ,[2] Joshi AV[2]
[1]RTI Health Solutions, Research Triangle Park, NC, [2]Novo Nordisk, Inc., Princeton, NJ

BACKGROUND

Nearly one in five civilian, non-institutionalized persons in the US sustains an injury each year that requires medical attention.[1] Previous research has estimated the economic burden associated with traumatic injury due specifically to blunt and penetrating mechanisms in a managed care population.[2] Few studies, though, have examined the inpatient costs and outcomes associated with blunt and penetrating trauma in detail (i.e., across all payers, by injury severity, by admitting facility's trauma center designation).

OBJECTIVE

Estimate per-discharge inpatient costs and outcomes for subjects with an admission for blunt or penetrating trauma, stratified by injury severity and admitting facility's trauma center designation.

METHODS

Study Design

Retrospective database analysis.

Data Source

Discharge data from the 2002 Healthcare Cost & Utilization Project (HCUP) Nationwide Inpatient Sample (NIS).

- NIS is the largest all-payer inpatient care database in the United States.
- The 2002 dataset contains data from approximately 7 million hospital stays.
- NIS is the only national hospital database with charge information on all patients, regardless of payer.
- NIS includes many clinical and non-clinical variables for each inpatient stay, including patient demographics, diagnosis codes, length of stay, total charges, admission and discharge status, payer, and hospital-specific characteristics.
- Sampling weights allow for nationally representative estimates.

Inclusion Criteria

- ≥1 ICD-9-CM diagnosis code consistent with blunt or penetrating trauma
 - Traumatic brain injury (TBI): ICD-9-CM codes 800–801.9, 803–804.9 or 850–854.1 regardless of mortality; 873.0–873.9, 905.0 or 907.0 for cases resulting in death (inclusive of all 4th and 5th digit modifiers, where appropriate).[3]
 - Other trauma: ICD-9-CM codes 802, 805–807, 808–809, 860–869 or 900–904 (inclusive of all 4th and 5th digit modifiers, where appropriate).
- Patients not admitted from, or discharged to, another inpatient facility.
 - Unique patient identifiers are not provided, so we were unable to follow patients who moved from facility to facility.

Analysis Strata

- Trauma type: (1) isolated TBI; (2) other trauma with TBI (T+TBI); and (3) other trauma without TBI (T-TBI).
- Injury severity: ICDMAP-90 software[4] used to assign Injury Severity Score (ISS).[5]
 - 1-9: Low
 - 10-15: Moderate
 - 16-24: Severe
 - 25+: Critical
- Trauma center designation[6] of admitting hospital: assigned using linked data from the American Hospital Association (AHA).
 - Level I: Has a full range of specialists and equipment available 24 hours a day and admits a minimum required annual volume of severely injured patients.
 - Level II: Provides comprehensive trauma care and supplements the clinical expertise of a Level I institution. Provides 24-hour availability of all essential specialties, personnel and equipment.
 - Level III: Does not have the full availability of specialists, but does have resources for the emergency resuscitation, surgery and intensive care of most trauma patients.
 - Level IV: Provides the stabilization and treatment of severely injured patients in remote areas where no alternative care is available.

Outcome Measures

- Per-discharge costs
 - NIS's charge data converted to costs using facility-specific cost-to-charge ratios
 - Costs converted to 2006 US dollars
- Per-discharge length of stay (LOS)
- Cost per day
- Probability of death

RESULTS

Prevalence (Table 1)

- 664,474 trauma-related discharges in 2002.
- Of these, 596,762 met all inclusion criteria; subsequent analyses were conducted on this population.
- Most admissions (64.3%) were for low-severity injuries (ISS=1-9).
- Critical injuries (ISS=25+) represented 6.4% of admissions.
- More than half (54.5%) of all admissions were to non-trauma centers.
- Level I, II, and III/IV trauma centers represented 20.5%, 20.2%, and 4.1% of admissions, respectively.

Table 1. Trauma-Related Discharges by Study Cohort, Trauma Center Level, and Injury Severity

	All Levels Combined	Level I		Level II		Level III/IV		Not a Trauma Center		Trauma Level Unknown	
	N	N	%	N	%	N	%	N	%	N	%
Isolated TBI											
All Severities Combined	136,836	32,493	100.00	33,539	100.00	3,978	100.00	65,580	100.00	1,245	100.00
ISS 1-9	61,028	13,778	42.40	16,602	49.50	2,268	57.02	27,792	42.38	588	47.21
ISS 10-15	7,940	2,364	7.28	1,923	5.73	250	6.29	3,301	5.03	102	8.15
ISS 16-24	56,215	13,165	40.52	12,245	36.51	1,247	31.34	29,114	44.40	444	35.62
ISS 25+	10,481	2,921	8.99	2,548	7.60	190	4.77	4,732	7.22	91	7.30
Unable to Score	1,171	265	0.82	221	0.66	23	0.57	641	0.98	21	1.72
Co-morbid TBI and Other Trauma											
All Severities Combined	63,078	21,458	100.00	15,314	100.00	1,460	100.00	24,279	100.00	566	100.00
ISS 1-9	11,119	3,272	15.25	3,125	20.41	404	27.68	4,238	17.45	80	14.15
ISS 10-15	13,995	4,325	20.15	3,404	22.23	470	32.22	5,657	23.30	139	24.53
ISS 16-24	20,510	7,279	33.92	4,691	30.63	414	28.37	7,965	32.80	160	28.30
ISS 25+	17,450	6,578	30.65	4,094	26.73	171	11.73	6,419	26.44	187	33.02
Unable to Score	5	5	0.02	0	0.00	0	0.00	0	0.00	0	0.00
Other Trauma Without Co-morbid TBI											
All Severities Combined	396,848	68,535	100.00	71,540	100.00	18,987	100.00	235,252	100.00	2,533	100.00
ISS 1-9	311,807	46,887	68.41	55,729	77.90	15,544	81.87	191,915	81.58	1,732	68.35
ISS 10-15	47,435	11,340	16.55	9,142	12.78	2,007	10.57	24,502	10.42	444	17.51
ISS 16-24	22,999	6,829	9.96	4,105	5.74	847	4.46	10,987	4.67	230	9.07
ISS 25+	10,163	3,012	4.39	1,906	2.66	327	1.72	4,812	2.05	107	4.22
Unable to Score	4,444	467	0.68	659	0.92	261	1.37	3,036	1.29	21	0.84
All Injuries Combined											
All Severities Combined	596,762	122,487	100.00	120,393	100.00	24,425	100.00	325,112	100.00	4,345	100.00
ISS 1-9	383,954	63,937	52.20	75,456	62.67	18,217	74.58	223,945	68.88	2,400	55.23
ISS 10-15	69,370	18,029	14.72	14,469	12.02	2,728	11.17	33,461	10.29	684	15.74
ISS 16-24	99,724	27,274	22.27	21,042	17.48	2,508	10.27	48,067	14.78	834	19.19
ISS 25+	38,094	12,511	10.21	8,547	7.10	688	2.82	15,963	4.91	385	8.86
Unable to Score	5,620	737	0.60	880	0.73	284	1.16	3,677	1.13	43	0.98

Patient Characteristics (Table 2)

- 136,836 (22.93%) had isolated TBI, 63,078 (10.57%) had T+TBI, and 396,848 (66.50%) had T-TBI.
- In T-TBI cohort, 78.76% had low-severity injuries (ISS 1-9).
- In T+TBI cohort, 60.18% had severe (ISS 16-24) or critical (ISS 25+) injuries.
- The T+TBI cohort had the highest proportion of admissions to a Level I or II trauma center (58.30%) and the lowest proportion admitted to a non-trauma center (38.49%).
- 54.48% of subjects were admitted to a non-trauma center.
- T+TBI cohort had youngest mean age (41.07 years), the greatest proportion of males (68.45%) and the highest mean ISS (19.58).

Table 2. Characteristics of the Study Sample

	Study Cohort							
	All Patients (N = 596,762; 100.00%)		Isolated TBI (N = 136,836; 22.93%)		T+TBI (N = 63,078; 10.57%)		T-TBI (N = 396,848; 66.50%)	
Characteristic	N	%	N	%	N	%	N	%
Age Category								
<20	75,295	12.62	29,078	21.25	11,138	17.66	35,079	8.84
20-39	139,215	23.33	30,107	22.00	22,462	35.61	86,646	21.83
40-64	142,632	23.90	30,779	22.49	18,130	28.74	93,723	23.62
65+	239,620	40.15	46,872	34.25	11,348	17.99	181,399	45.71
Mean Age (Years)	52.80		47.33		41.07		56.55	
Gender								
Female	253,208	42.43	52,230	38.17	19,904	31.55	181,074	45.63
Male	343,524	57.56	84,590	61.82	43,174	68.45	215,760	54.37
Missing	31	0.01	16	0.01	0	0	15	0.00
Injury Severity Score (ISS)								
1-9	383,954	64.34	61,028	44.60	11,119	17.63	311,807	78.57
10-15	69,370	11.62	7,940	5.80	13,995	22.19	47,435	11.95
16-24	99,724	16.71	56,215	41.08	20,510	32.52	22,999	5.80
25+	38,094	6.38	10,481	7.66	17,450	27.66	10,163	2.56
Unable to Score	5,620	0.94	1,171	0.86	5	0.01	4,444	1.12
Mean ISS	9.85		12.00		19.58		7.55	
Trauma Level of Admitting Hospital								
I	122,487	20.53	32,493	23.75	21,458	34.02	68,535	17.27
II	120,393	20.17	33,539	24.51	15,314	24.28	71,540	18.03
III/IV	24,425	4.09	3,978	2.91	1,460	2.31	18,987	4.78
Trauma Ctr, but Level Unknown	4,345	0.73	1,245	0.91	566	0.90	2,533	0.64
Not a Trauma Center	325,112	54.48	65,580	47.93	24,279	38.49	235,252	59.28

RESULTS (continued)

Per-discharge total costs, length of stay, cost-per-day and probability of death (Figures 1 and 2)

- In T+TBI with critical (ISS 25+) injuries, total costs were nearly double ($51,603) those incurred for isolated TBI ($26,600).
- The longest LOS was found in the T+TBI cohort with critical injuries, while the shortest was found in the isolated TBI cohort with low-severity injuries.
- The rate of death was highest (nearly 50%) among the isolated TBI cohort with critical injuries; it was nearly twice the rate for the T+TBI group with critical injuries.
- Within study cohorts, but across injury severities, the cost-per-day did not vary greatly.
- Across all study cohorts, total costs were greater for patients in Level I trauma centers.
- Across all study cohorts, LOS was shortest among those in Level III/IV trauma centers but did not vary across Level I, Level II and non-trauma facilities.
- Within study cohorts, the probability of death did not vary greatly across trauma center levels.

Figure 1. Total costs, length of stay, cost-per-day and probability of death by study cohort and injury severity score (ISS).

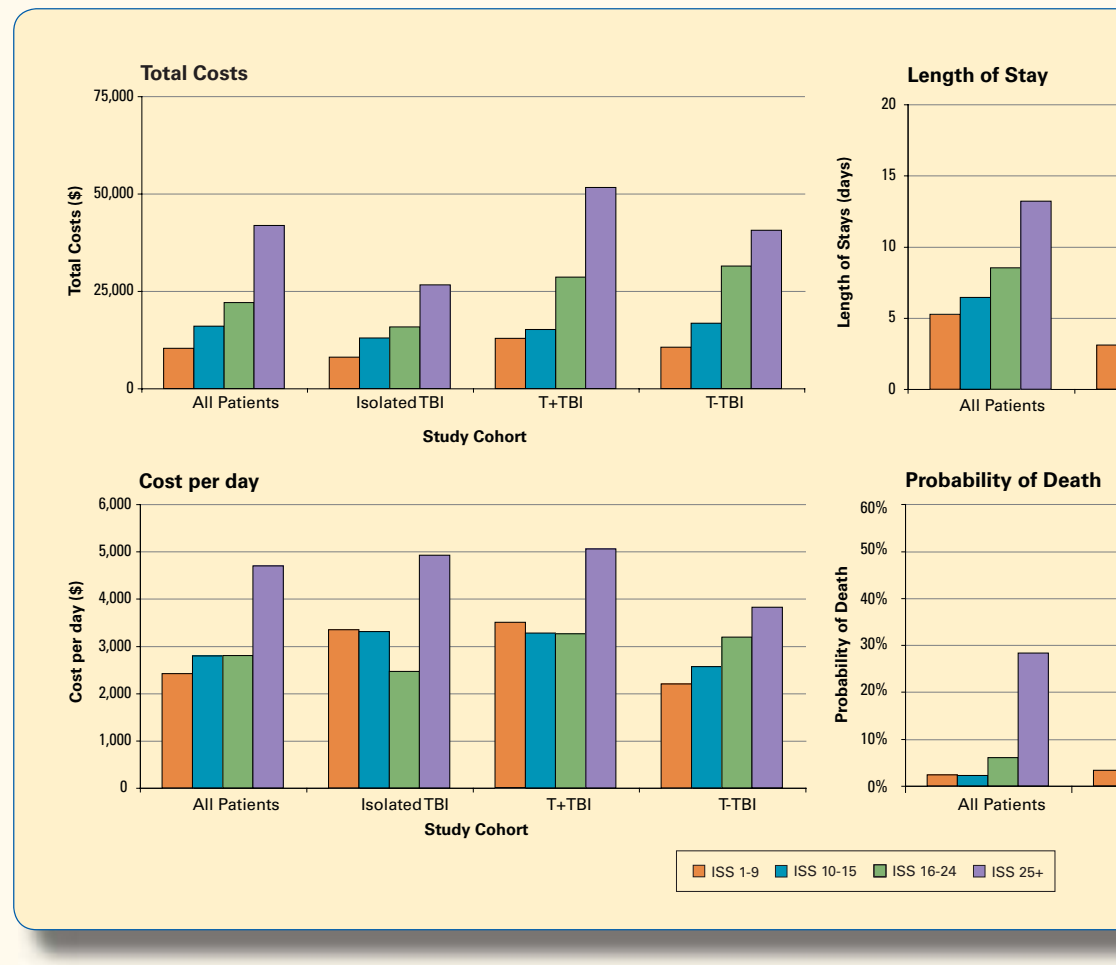
Total Costs
Total Costs ($)
Length of Stay
Length of Stays (days)
Cost per day
Cost per day ($)
Probability of Death
All Patients
Isolated TBI
T+TBI
T-TBI
Study Cohort
ISS 1-9
ISS 10-15
ISS 16-24
ISS 25+

Figure 2. Total costs, length of stay, cost-per-day and probability of death by study cohort and trauma center level of the admitting hospital.

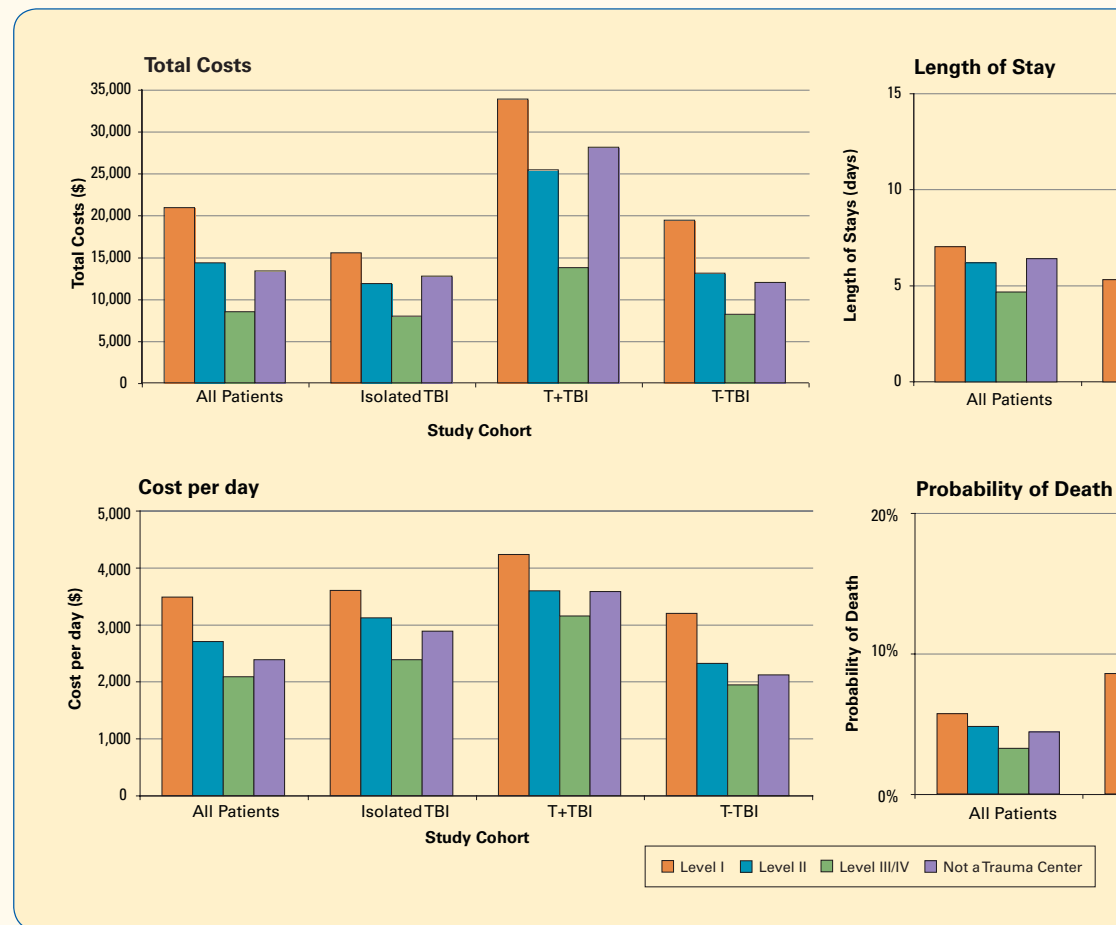
Total Costs
Total Costs ($)
Length of Stay
Length of Stays (days)
Cost per day
Cost per day ($)
Probability of Death
All Patients
Isolated TBI
T+TBI
T-TBI
Study Cohort
Level I
Level II
Level III/IV
Not a Trauma Center

LIMITATIONS

- Subjects were identified based on diagnosis codes, which, if recorded inaccurately, may cause some patients to be misidentified as having trauma or to be misclassified by ISS.
- Discharge status and external injury cause codes (E-codes) may be unreliably recorded in these data; therefore, we are unable to provide context with regard to these parameters.
- Because unique patient identifiers are not provided, we were unable to follow patients who move from facility to facility. Results may be biased somewhat if the experiences of patients who transferred from facility to facility differ from those who remained in the analytic sample.

CONCLUSIONS

- The economic burden of blunt and penetrating trauma in hospitalized patients is substantial.
- Results highlight the substantially higher costs, LOS, and probability of death associated with combined TBI and other trauma (T+TBI).
- Specialized Level I trauma centers have higher costs.
- TBI contributes dramatically to trauma patients' injury severity, whether alone or in combination with other trauma.
- Results provide insight regarding the degree to which trauma systems are functioning.
- Regardless of injury severity, many patients are admitted to non-trauma centers.

REFERENCES

1. Finkelstein EA, Corso PS, Miller TR. The Incidence and Economic Burden of Injuries in the United States. New York, NY: Oxford University Press; 2006.
2. Davis KL, Joshi AV, Tortella BJ, Candrilli SD. The direct economic burden of blunt and penetrating trauma in a managed care population. J Trauma. 2007 Mar; 62(3):622-30.
3. Thurman DJ, Sniezek JE, Johnson D, et al. Guidelines for Surveillance of Central Nervous System Injury. Atlanta, GA: Centers for Disease Control and Prevention (CDC); 1995.
4. The Johns Hopkins University and Tri-Analytics, Inc. ICDMAP-90 (computer software). Baltimore, MD; 1998.
5. MacKenzie EJ, Steinwachs DM, Shankar B. Classifying trauma severity based on hospital discharge diagnoses: validation of an ICD-9-CM to AIS-85 conversion table. Med Care. 1989 Apr;27:412-22.
6. MacKenzie EJ, Hoyt DB, Sacra JC, et al. National Inventory of Hospital Trauma Centers. JAMA. 2003 Mar;289(12):1515-22.

ACKNOWLEDGMENT

This study was funded in part by Novo Nordisk, Inc.

CONTACT INFORMATION

Sean Candrilli, MS
Senior Director, Health Economics

RTI Health Solutions, RTI International
3040 Cornwallis Road, PO Box 12194
Research Triangle Park, NC 27709-2194

Phone: 412.421.1578
E-mail: scandrilli@rti.org

Presented at:
12th Annual International Meeting of the International Society for Pharmacoeconomics and Outcomes Research
May 19-23, 2007, Arlington, VA